Orforglipron for Maintenance of Body Weight Reduction: The Double-Blind, Randomized, Phase 3b ATTAIN-MAINTAIN Trial

Lilly
A MEDICINE COMPANY

Orforglipron is not FDA-approved to maintain weight reduction long term after switching from injectable incretins (tirzepatide or semaglutide) in adults with obesity or in adults with overweight with at least one weight-related comorbid condition. Orforglipron is not FDA-approved for initiation at the 9 mg dose. The safety and effectiveness of these unapproved uses have not been established.

This information is intended for US HCPs for medical, scientific, and educational purposes. Not intended for product promotion.

Orforglipron is a GLP-1 receptor agonist indicated, in combination with a reduced-calorie diet and increased physical activity, to reduce excess body weight and maintain weight reduction long term in adults with obesity or in adults with overweight with at least one weight-related comorbid condition. Concomitant use with another GLP-1 receptor agonist is not recommended.

This study was supported by Eli Lilly and Company.

The following authors, editors, and contributors to the publication were employees of, consultants to, or received compensation from Lilly: Louis J. Aronne, Deborah B. Horn, Carel W. Le Roux, Ariana M. Chao, Wayne Ho, Bruno Halpern, Ryan Griffin, Cathy Xie, Elisa Gomez Valderas, Clare J. Lee, Anderson Ribeiro, David M. Hyman, Leonard Glass, Neena Xavier

Limitations of this analysis of ATTAIN-MAINTAIN:

- Study sites were limited to the USA with a predominantly White study population, although the study includes a more representative US population with a higher proportion of Black or African/ American and Hispanic/Latino population
- Additional trial limitations include the absence of a comparator arm involving continued use of injectable obesity-management medications and the trial's 1-year duration
- Due to the increasing availability of OMMs, there were participants who discontinued treatment to start available and approved medications, even before the 24 weeks when rescue therapy became an option for the placebo

Click here to access the full Prescribing Information for orforglipron.

WARNING: RISK OF THYROID C-CELL TUMORS
See full prescribing information for complete boxed warning.

- In products with GLP-1 receptor agonist activity that are pharmacologically active in rats and mice, rodent thyroid C-cell tumors (adenomas and carcinomas) have been observed at clinically relevant exposures and are considered GLP-1 receptor-dependent effects in rodents. Orforglipron is not pharmacologically active in rats or mice and did not produce tumors in rodents (13.1). While orforglipron is pharmacologically active at the human GLP-1 receptor, the human relevance of GLP-1 receptor-dependent thyroid C-cell tumors observed in rodents has not been determined (5.1, 13.1)
- Orforglipron is contraindicated in patients with a personal or family history of MTC or in patients with MEN 2. Counsel patients regarding the potential risk of MTC and symptoms of thyroid tumors (4, 5.1)

Contraindication:

Orforglipron is contraindicated in patients with a personal or family history of MTC or in patients with MEN 2 and patients with known serious hypersensitivity to orforglipron or any of the excipients in orforglipron.

Highlights of Prescribing Information | Warnings and Precautions:

- Acute Pancreatitis: Has been observed in patients treated with GLP-1 receptor agonists, including orforglipron. Discontinue if pancreatitis is suspected (5.2)
- Severe Gastrointestinal Reactions: Use of orforglipron has been associated with gastrointestinal adverse reactions, sometimes severe. Orforglipron is not recommended in patients with severe gastroparesis. (5.3)
- Acute Kidney Injury Due to Volume Depletion: Monitor renal function in patients reporting adverse reactions that could lead to volume depletion (5.4)
- Hypoglycemia: Concomitant use with insulin or an insulin secretagogue may increase the risk of hypoglycemia, including severe hypoglycemia. Reducing the dosage of insulin or insulin secretagogue may be necessary. Inform all patients of the risk of hypoglycemia and educate them on the signs and symptoms of hypoglycemia (5.5)
- Hypersensitivity Reactions: Serious hypersensitivity reactions (eg, anaphylaxis, angioedema) have been reported with GLP-1 receptor agonists. If suspected, advise the patient to promptly seek medical attention and discontinue orforglipron (5.6)
- Diabetic Retinopathy Complications in Patients With Type 2 Diabetes: Orforglipron has not been studied in patients with diabetic retinopathy and/or macular edema requiring acute treatment. Monitor patients with a history of diabetic retinopathy for progression (5.7)
- Acute Gallbladder Disease: Has been reported in clinical trials. If cholecystitis is suspected, gallbladder studies and clinical follow-up are indicated (5.8)
- Pulmonary Aspiration During General Anesthesia and Deep Sedation: Has been reported in patients receiving GLP-1 receptor agonists undergoing elective surgeries or procedures. Instruct patients to inform healthcare providers of any planned surgeries or procedures (5.9)

Click below to see the full results published in *Nature Medicine*

ATTAIN-MAINTAIN

FDA=US Food and Drug Administration; GLP-1=glucagon-like peptide-1; HCP=healthcare professional; MEN 2=multiple endocrine neoplasia syndrome type 2; MTC=medullary thyroid carcinoma; OMM=obesity management medication.

MMAT-04107 © 2026 Eli Lilly and Company. All rights reserved.